Discriminant validity of the Work Productivity and Activity Impairment questionnaire in patients with Crohn's disease

Reilly MC,[1] Gerlier L,[2] Brown M[3]

[1]Margaret Reilly Associates, Inc, New York, NY, USA; [2]Keyrus Biopharma, Levallois-Perret, France; [3]UCB, Slough, UK

Background

- Crohn's disease (CD) is a chronic inflammatory disease of the gastrointestinal tract characterised by debilitating symptoms, including abdominal pain, weight loss, anaemia and persistent diarrhoea. It emerges in early adulthood and has no known cure. Patients with CD experience an ongoing cycle of remissions and unpredictable exacerbations, leading to a considerable degree of uncertainty in their lives.
- The efficacy and safety of certolizumab pegol, a PEGylated Fab' fragment of a humanised anti-TNF monoclonal antibody that neutralises tumour necrosis factor (TNF), has been demonstrated in the PRECiSE programme. PRECiSE 1[1] and PRECiSE 2[2] were large randomised controlled Phase III trials including over 1300 patients with moderate to severe CD, representing the largest and most comprehensive development programme for an anti-TNF in the treatment of CD.

Introduction

- The huge economic burden of CD is driven by hospitalisations and drug therapies, but more importantly by indirect costs: more than 70% of the total cost to society of CD is due to absences from work and losses of productivity.[3]
- The Work Productivity and Activity Impairment questionnaire[4] (WPAI) is used to collect data on the impairment due to a health problem during the previous 7 days. Results are summarised into four impairment scores expressed in percentages (%):
 - — Absenteeism: hours missed from work
 - — Presenteeism: impairment while working
 - — Overall work impairment: combination of Absenteeism and Presenteeism
 - — Daily activities impairment: eg shopping, exercising, childcare, etc.
- The validity of the WPAI questionnaire in the measurement of health related productivity was demonstrated in several diseases, including hepatitis C[5] and the digestive pathologies irritable bowel syndrome[6] and gastro-oesophageal reflux disease.[7]
- The CD-specific WPAI (WPAI:CD) was administered for the first time in the PRECiSE 1 and PRECiSE 2 studies, the baseline results of which already confirmed the reduced productivity of patients with active CD at work and in daily activities.[8]

Objectives

- The objective was to compare the WPAI:CD scores of patients categorised by baseline disease severity and health-related quality of life (HRQoL) level, with the a priori assumption that more severe health conditions are associated with higher impairments in work productivity and daily activities.

Methods

- The 1330 randomised patients were divided into 'best' and 'worst' subgroups determined by the CD Activity Index (CDAI), Medical Outcomes Short-Form (36-item) Health Survey (SF-36) and EuroQoL 5-Dimensional (EQ-5D) instruments.
- 'Best' health was identified by CDAI scores less than or equal to the median, SF-36 Physical and Mental Component Summary scores (PCS and MCS) greater than the median (Table 1) and modality '1' responses to the EQ-5D (Table 2).
- The baseline WPAI:CD scores (Table 3) were then calculated by subgroups and compared using a nonparametric Wilcoxon test.

Table 1. Thresholds used in the analysis for CDAI, SF-36 PCS and MCS (PRECiSE 1 and 2, baseline, all subjects)

Instrument	n	Median score
CDAI[a]	1322	288.1
SF-36 Physical Component Summary[b]	1304	37.5
SF-36 Mental Component Summary[b]	1304	37.4

[a]Lower is better. [b]Higher is better.

Table 2. Proportion of patients without problems, as defined by the EQ-5D for each dimension (PRECiSE 1 and 2, baseline, all subjects)

EQ-5D dimension	n	Proportion of patients with 'No problems'[a] at baseline (%)
Mobility	1324	62.7
Self-care	1324	93.1
Usual activities	1323	26.0
Pain/discomfort	1324	4.3
Anxiety/depression	1325	30.2

[a]I.e. selecting modality '1'.

Table 3. WPAI:CD results (PRECiSE 1 and 2, baseline, all subjects)

WPAI:CD dimension	n	Mean (SD) impairment (%)[a]
Absenteeism	706	19.5 (29.2)
Presenteeism	715	42.0 (24.8)
Overall work	656	47.5 (26.4)
Daily activities	1289	53.3 (24.9)

[a]Lower is better. SD = standard deviation.

Results

- CD patients with the 'worst' disease severity showed higher impairment in overall work (+8.9%) and activities (+9.6%) versus patients with 'best' health (Figure 1).

Figure 1. WPAI:CD scores by health state severity, as defined by CDAI baseline score (PRECiSE 1 and 2, all subjects).

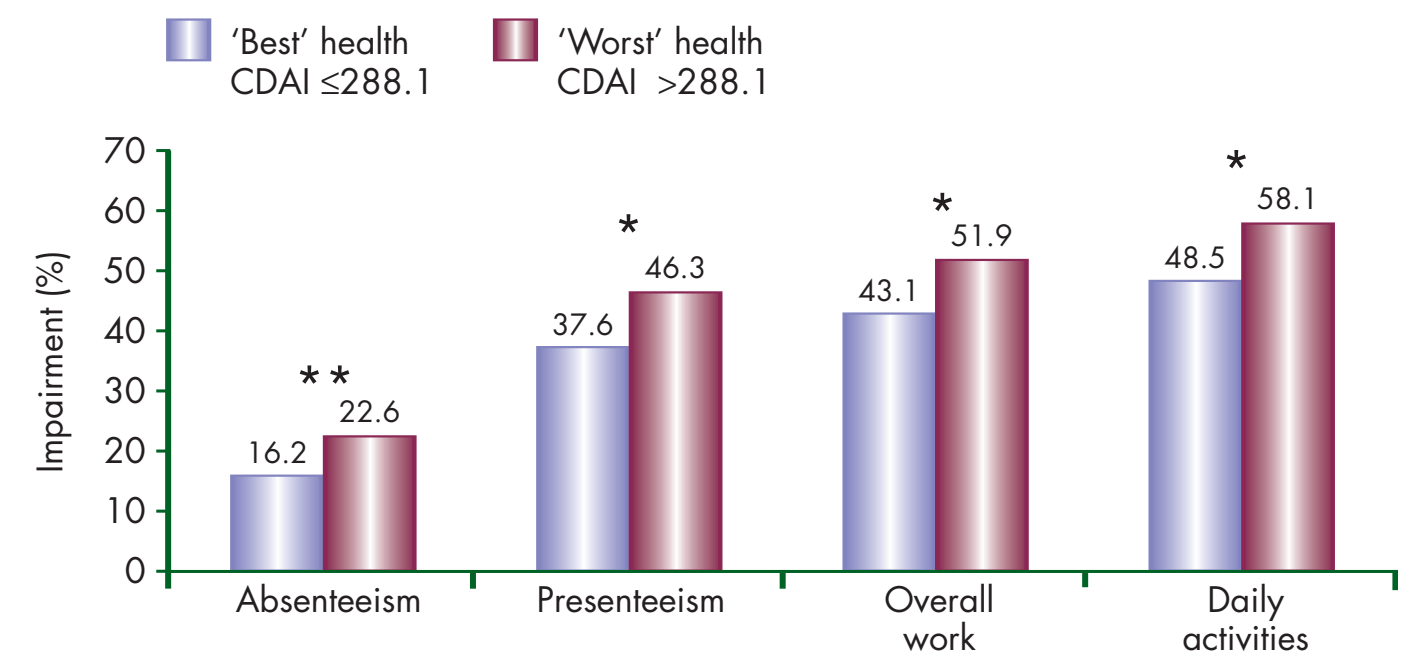
'Best' health CDAI ≤288.1
'Worst' health CDAI >288.1
Impairment (%)
70
60
50
40
30
20
10
0
16.2
**
22.6
37.6
*
46.3
43.1
*
51.9
48.5
*
58.1
Absenteeism
Presenteeism
Overall work
Daily activities

*p<0.001. **p=0.004.

- Patients with 'worst' HRQoL showed higher impairments in overall work (+20.0% by PCS; +18.5% by MCS) and activities (+22.2% by PCS; +15.5% by MCS) versus 'best' HRQoL (Figures 2 and 3).

Figure 2. WPAI:CD scores by health state severity, as defined by the SF-36 Physical Component Summary score (PRECiSE 1 and 2, all subjects).

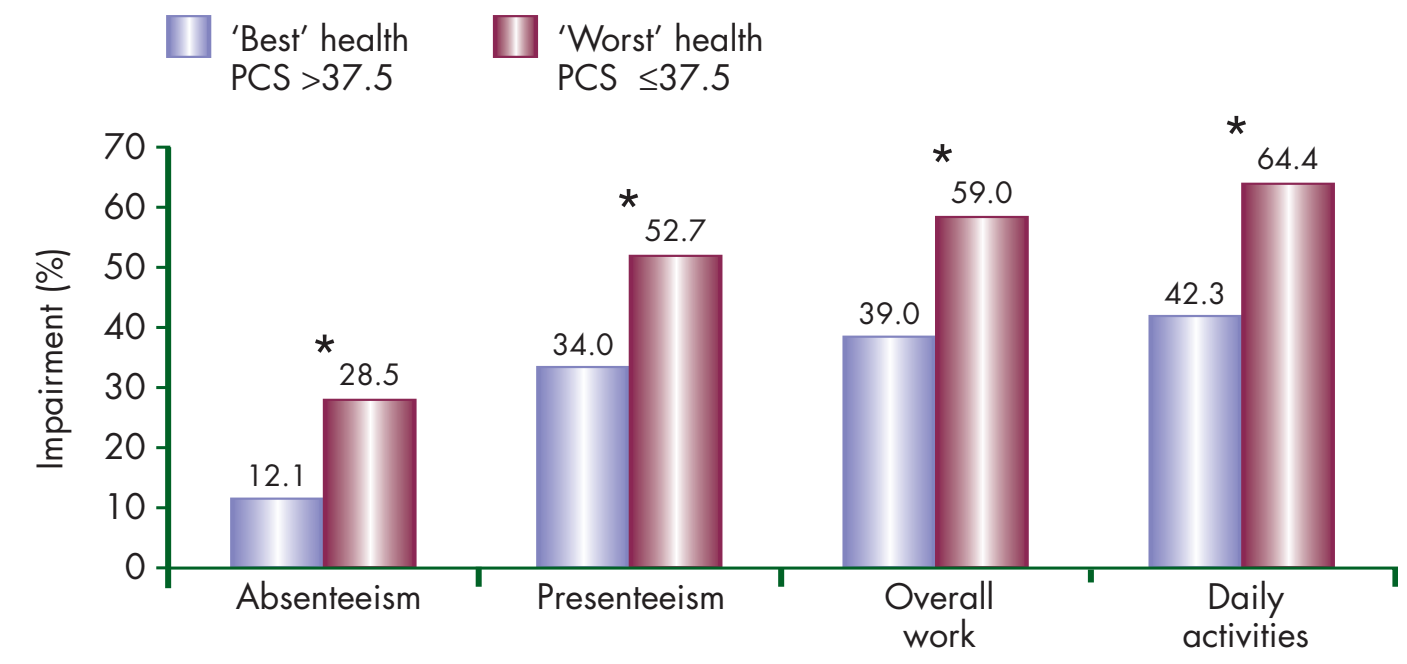
'Best' health PCS >37.5
'Worst' health PCS ≤37.5
Impairment (%)
70
60
50
40
30
20
10
0
12.1
*
28.5
34.0
*
52.7
39.0
*
59.0
42.3
*
64.4
Absenteeism
Presenteeism
Overall work
Daily activities

*p<0.001.

Figure 3. WPAI:CD scores by health state severity, as defined by the SF-36 Mental Component Summary score (PRECiSE 1 and 2, all subjects).

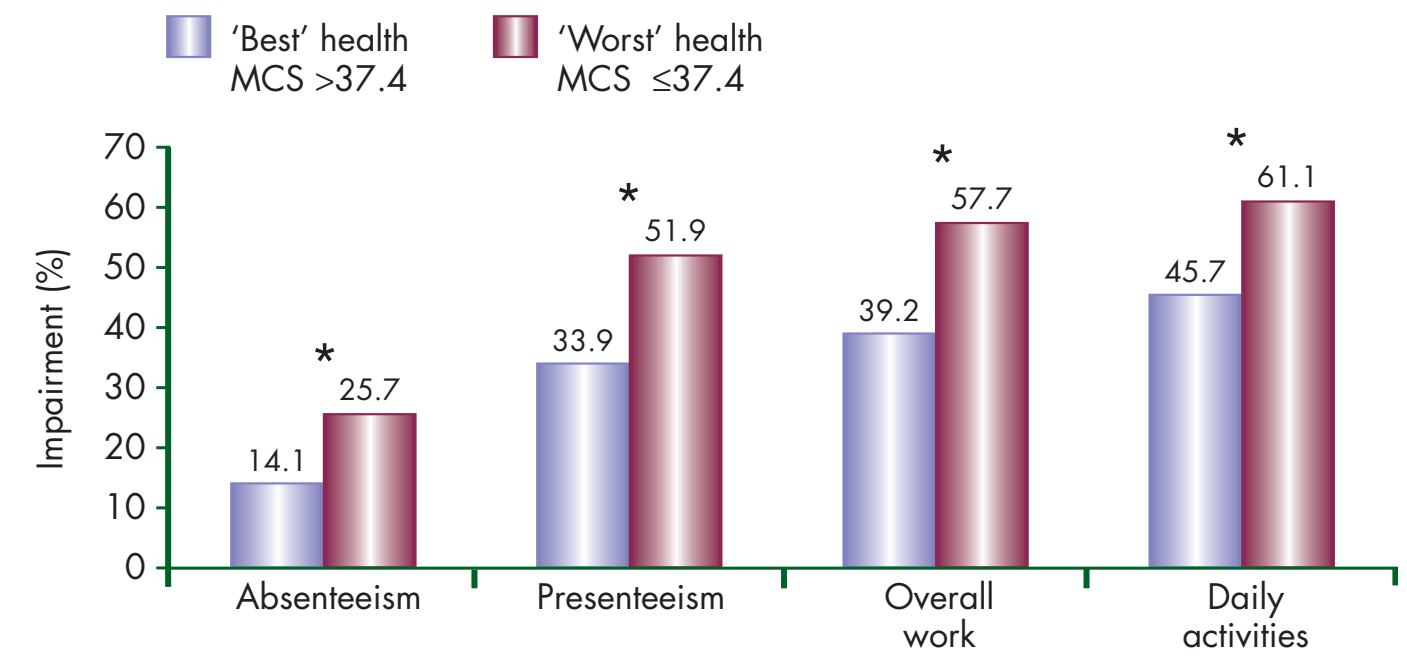
'Best' health MCS >37.4
'Worst' health MCS ≤37.4
Impairment (%)
70
60
50
40
30
20
10
0
14.1
*
25.7
33.9
*
51.9
39.2
*
57.7
45.7
*
61.1
Absenteeism
Presenteeism
Overall work
Daily activities

*p<0.001.

- Patients with the 'worst' EQ-5D states reported more impairment in overall work (+15.2% averaged over the five dimensions) and activities (+17.3%) versus patients with the 'best' states (Figure 4).

Figure 4. Differences in impairment between 'worst' and 'best' health states defined by the EQ-5D answers, Overall work and Daily activities dimensions (PRECiSE 1 and 2, all subjects).

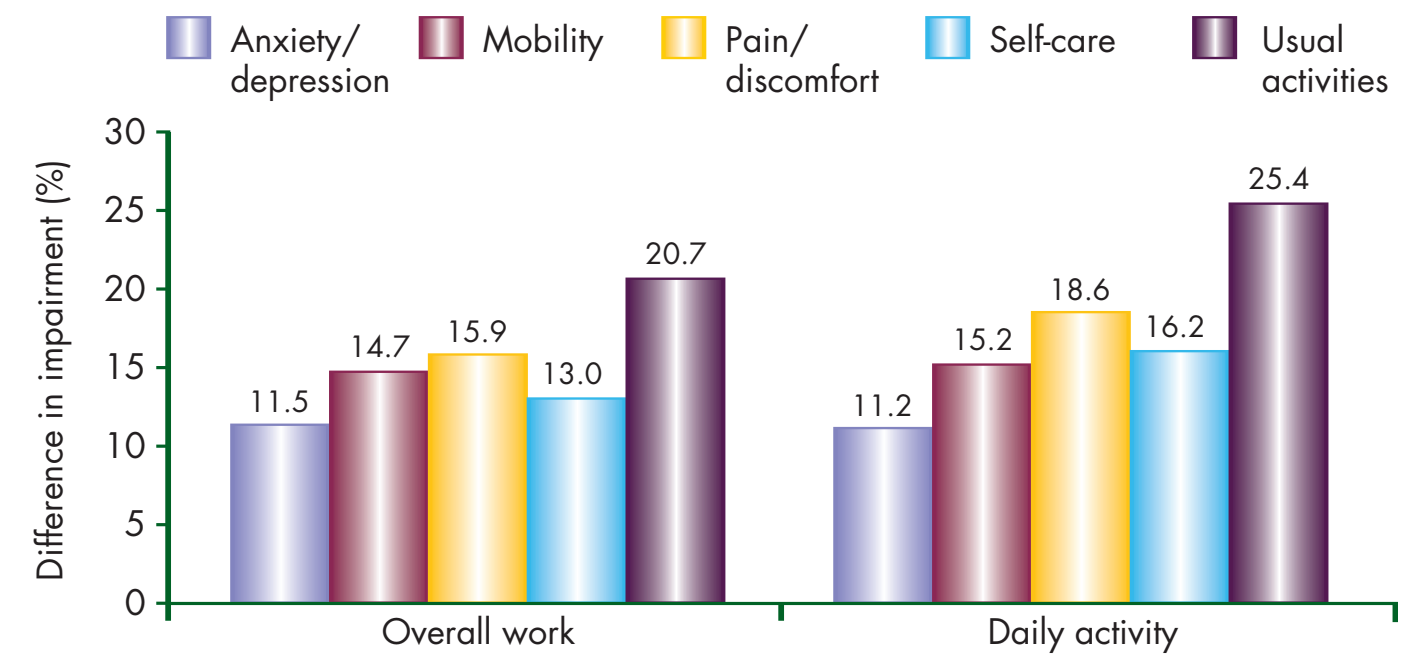
Anxiety/ depression
Mobility
Pain/ discomfort
Self-care
Usual activities
Difference in impairment (%)
30
25
20
15
10
5
0
11.5
14.7
15.9
13.0
20.7
11.2
15.2
18.6
16.2
25.4
Overall work
Daily activity

p-values varied from <0.001 to 0.009. Only one p-value was nonsignificant: Self-care dimension, Presenteeism.

Conclusion

- The measurement of expected differences in work and activity impairment by disease severity and HRQoL levels supports the discriminant validity of the WPAI:CD.
- The WPAI:CD is a valid tool for assessing the indirect costs of CD.

References

1. Schreiber S, et al. Gut 2005;54(Suppl. VII):A82(Abstract).
2. Sandborn WJ, et al. Gastroenterology 2006;130(Suppl. 2):A-107.
3. Ekbom A, et al. Research and Clinical Forums 1998;20:33–39.
4. Reilly MC, et al. Pharmacoeconomics 1993;4:353–365.
5. Perrillo R, et al. J Viral Hepat 2004;11:157–165.
6. Reilly MC, et al. Aliment Pharmacol Ther 2004;20:459–467.
7. Wahlqvist P, et al. Value Health 2002;5:106–113.
8. Feagan BG, et al. Gastroenterology 2006;130(Suppl. 2):A-483.