Correlation between different productivity variables obtained from the WPAI-GERD questionnaire

Wahlqvist P, Carlsson J

AstraZeneca R&D Mölndal, Sweden

Conclusions

Results obtained from the WPAI-GERD in the current study indicated that answers were given in a consistent manner and according to expectations, thus lending support to construct validity of the questionnaire.

Background

Based on results from a cross-sectional study of a Swedish working population (N=136) visiting a general practitioner due to Gastro-Esophageal Reflux Disease (GERD), a validation analysis of the Work Productivity and Activity Impairment questionnaire (WPAI) for assessing lost productivity due to symptoms of GERD has previously been carried out and reported[1]. The construct validity of the WPAI-GERD was found to be high, and patients with symptoms of GERD (heartburn) reported an average of 2.5 hours absence from work, 23% reduced productivity while at work and 30% reduced productivity while doing regular daily activities during the week preceding the consultation. Loss of productivity due to GERD symptoms was found to increase with the severity of heartburn (Figure 1).

Results obtained from the WPAI questionnaire cannot be used to assess internal consistency reliability[2], since the different productivity variables by definition do not measure the same construct. However, evidence of answers being provided in a consistent manner and according to expectations would lend further support to construct validity of the questionnaire.

Objectives

To carry out a further validation analysis of the WPAI-GERD by investigating the relationship between the different productivity variables obtained (GERD-related absence from work, reduced productivity while at work, and reduced productivity while doing regular daily activities) in a previously reported study.

Methods

Pearson correlation coefficients between each productivity variable were calculated based on results from the original study population (including 27 patients without heartburn symptoms). Correlations were also assessed for each category of symptom severity in patients with heartburn (mild, n=30; moderate, n=64; severe, n=15).

Results

The correlation between reduced productivity while at work and reduced productivity while doing regular daily activities was found to be high and statistically significant (Table 1). This finding was consistent in patients with either mild, moderate or severe heartburn.

The proportion of patients reporting any absence from work was higher the more severe symptoms patients had (mild: 7% (2/30), moderate: 28% (18/64), severe: 33% (5/15)), and only two patients with mild heartburn reported any absence from work because of GERD. Despite the fact that the number of patients also with moderate or severe heartburn reporting absence from work is relatively small as a basis for analysis, a positive relationship was found between absence from work and reduced productivity at work, as well as between absence from work and reduced productivity in daily activities. Hence, results indicate that absence from work because of symptoms of GERD is also associated with reduced productivity while at work and while doing regular daily activities.

References

1. Wahlqvist P, Carlsson J, Stålhammar N-O, Wiklund I. Validity of a Work Productivity and Activity Impairment Questionnaire for Patients with Symptoms of Gastro-Esophageal Reflux Disease (WPAI-GERD) - Results From a Cross-Sectional Study. *Value in Health* 2002;5:106-113.
2. Bowling A. Theory of measurement. Chapter 2 in *Measuring health - a review of quality of life measurement scales.* Buckingham; Open University Press, 1998.

Figure 1. Loss of productivity by severity of heartburn (none, n=27; mild, n=30; moderate, n=64; severe, n=15), mean values per patient.

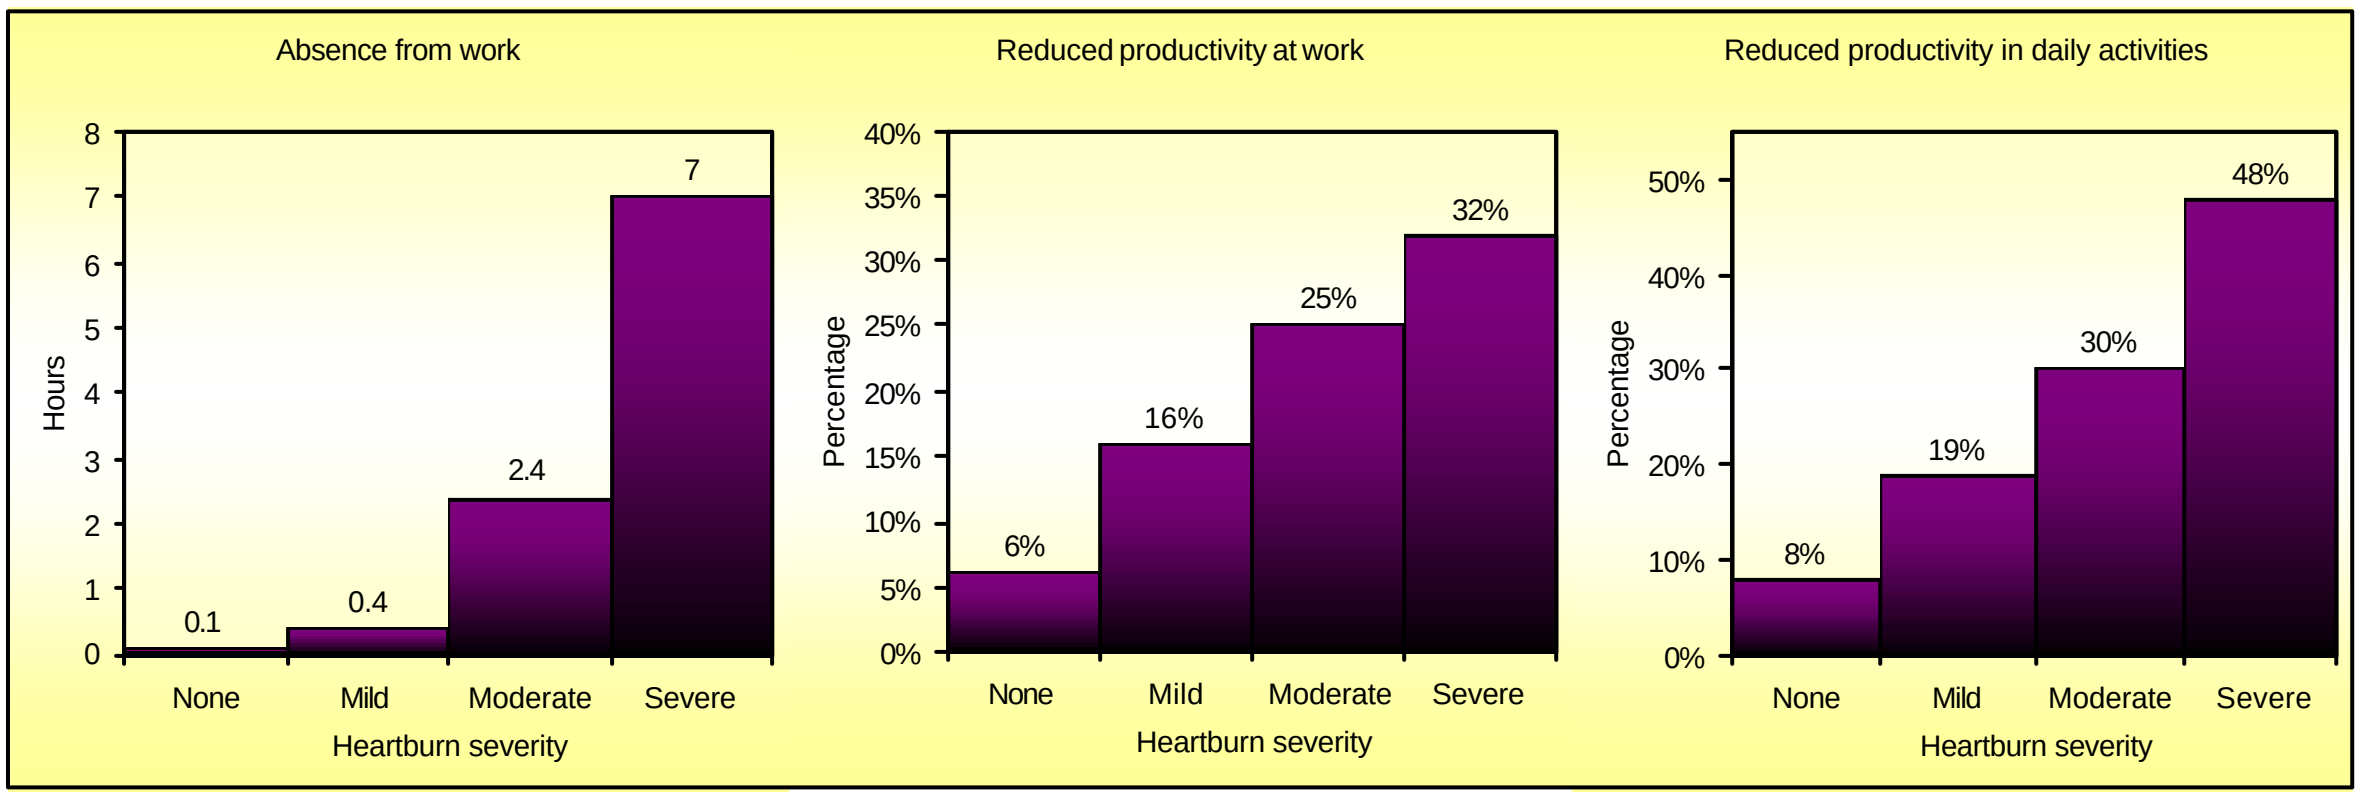

Absence from work
Hours
0.1
0.4
2.4
7
None
Mild
Moderate
Severe
Heartburn severity
Reduced productivity at work
Percentage
6%
16%
25%
32%
None
Mild
Moderate
Severe
Heartburn severity
Reduced productivity in daily activities
Percentage
8%
19%
30%
48%
None
Mild
Moderate
Severe
Heartburn severity

Table 1. Pearson correlation coefficients.

	All patients (N=136)		Mild heartburn (N=30)		Moderate heartburn (N=64)		Severe heartburn (N=15)	
	PW	PA	PW	PA	PW	PA	PW	PA
AW	0.57*	0.52*	-0.19	0.02	0.48*	0.39**	0.78**	0.73**
PW	-	0.82*	-	0.77*	-	0.79*	-	0.77**

* p<0.0001, ** p<0.002, AW = Absence from Work, PW = average reduced Productivity while Working, PA = average reduced Productivity while doing regular daily Activities.